Localizing Event-Related Potentials using New Approaches to Multi-source Minimum Variance Beamforming

Anthony Herdman[1], Alexander Moiseev[2], Urs Ribary[2]; [1]University of British Columbia, Canada, [2]Simon Fraser University, Canada

Introduction

- Adaptive and non-adaptive beamformers have become prominent neuroimaging tools for localizing neural sources of EEG and MEG data
- Validation of these methods using simulated data is required to improve our confidence when interpreting results obtained from real recordings
- Improvements in beamformer methods continue to advance our ability to localize the spatiotemporal dynamics of brain function
- Single-source beamformers (e.g., SPA) can miss or inaccurately localize highly-correlated (r>.8) sources (e.g., Moiseev et al., 2011; Van Veen et al., 1997)
- Multi-source beamformers (e.g., SIA & MIA) can improve localization performance and accuracy (e.g., Diwakar et al., 2011; Moiseev et al., 2011)
- More accurate source localization leads to more accurate source waveform reconstruction; thus, better interpretations of spatiotemporal dynamics
- Source mixing (a.k.a. leakage) can generate false connectivity patterns
- Multi-source null-constraints can significantly reduce or eliminate source mixing

Methods

Source Simulation / Source Reconstruction

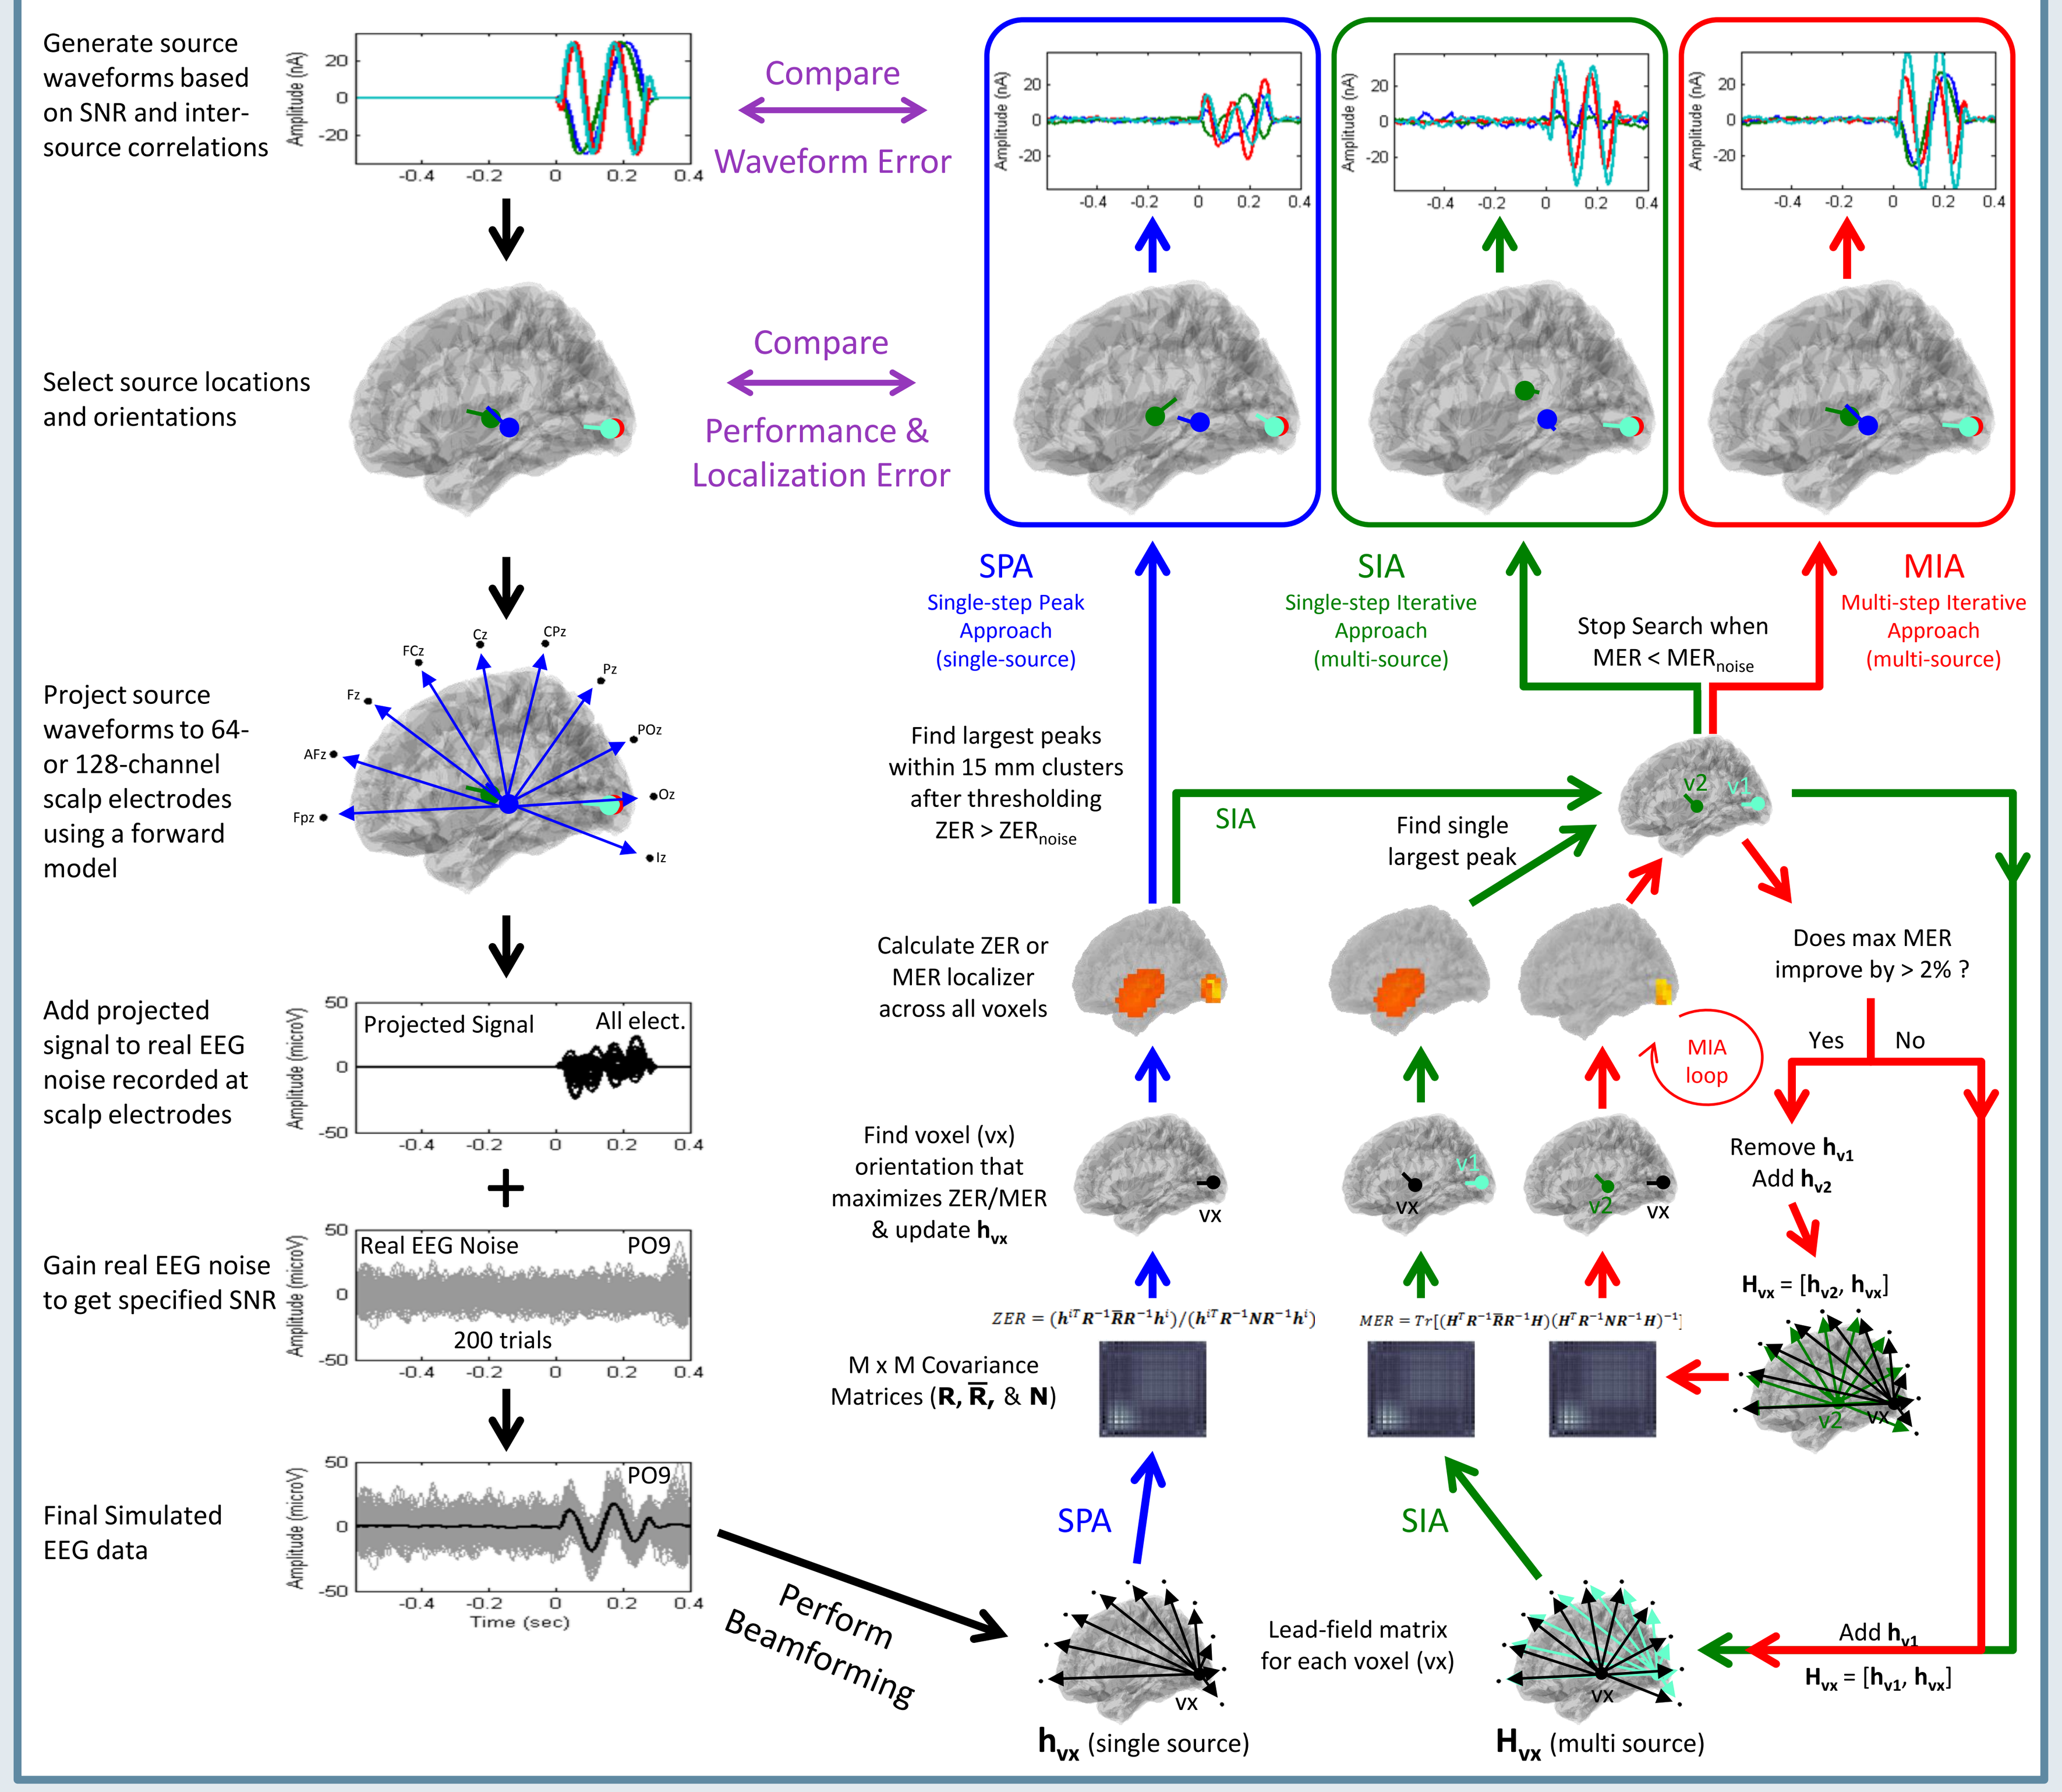
Generate source waveforms based on SNR and inter-source correlations
Select source locations and orientations
Project source waveforms to 64- or 128-channel scalp electrodes using a forward model
Add projected signal to real EEG noise recorded at scalp electrodes
Projected Signal
All elect.
Gain real EEG noise to get specified SNR
Real EEG Noise
PO9
200 trials
Final Simulated EEG data
Time (sec)
Compare
Waveform Error
Compare
Performance & Localization Error
Perform Beamforming
SPA
Single-step Peak Approach (single-source)
SIA
Single-step Iterative Approach (multi-source)
MIA
Multi-step Iterative Approach (multi-source)
Stop Search when MER < MER_noise
Find largest peaks within 15 mm clusters after thresholding ZER > ZER_noise
Find single largest peak
Calculate ZER or MER localizer across all voxels
Does max MER improve by > 2% ?
Yes
No
MIA loop
Find voxel (vx) orientation that maximizes ZER/MER & update h_vx
Remove h_v1
Add h_v2
H_vx = [h_v2, h_vx]
M x M Covariance Matrices (R, R̄, & N)
Lead-field matrix for each voxel (vx)
h_vx (single source)
H_vx (multi source)
Add h_v1
H_vx = [h_v1, h_vx]

$ZER = (h^{iT}R^{-1}\bar{R}R^{-1}h^{i})/(h^{iT}R^{-1}NR^{-1}h^{i})$

$MER = Tr[(H^{T}R^{-1}\bar{R}R^{-1}H)(H^{T}R^{-1}NR^{-1}H)^{-1}]$

Localization Results

Multi-source beamformers (MIA & SIA) outperformed Single-source beamformer (SPA)

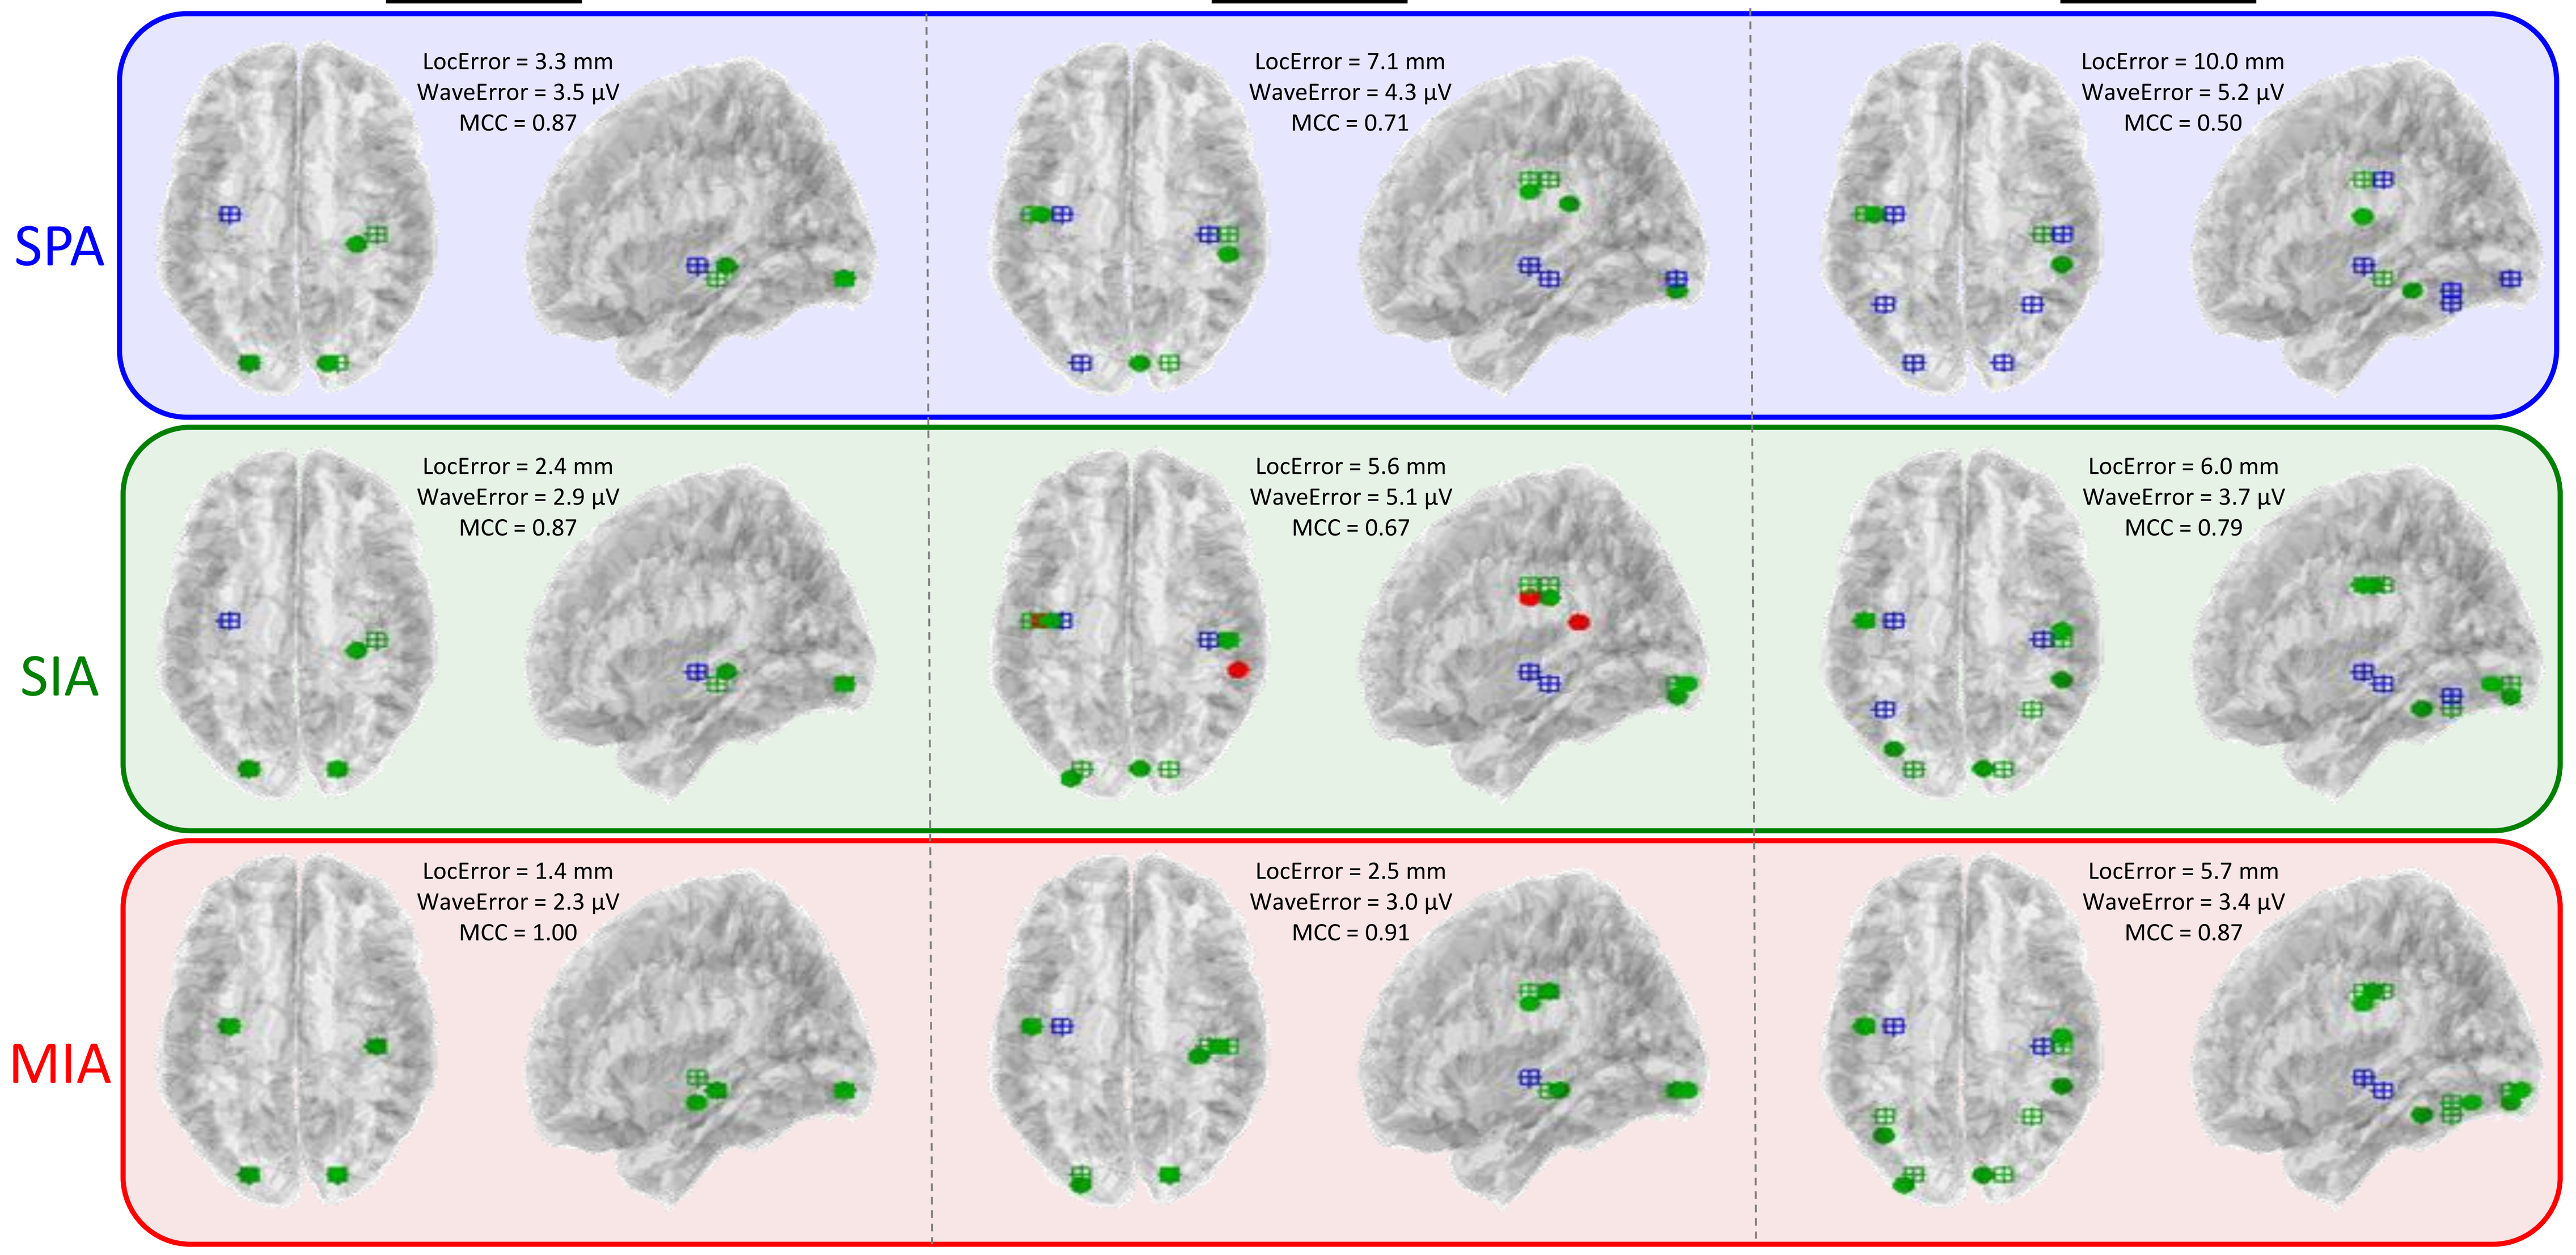
4 sources
6 sources
8 sources
SPA
LocError = 3.3 mm
WaveError = 3.5 µV
MCC = 0.87
LocError = 7.1 mm
WaveError = 4.3 µV
MCC = 0.71
LocError = 10.0 mm
WaveError = 5.2 µV
MCC = 0.50
SIA
LocError = 2.4 mm
WaveError = 2.9 µV
MCC = 0.87
LocError = 5.6 mm
WaveError = 5.1 µV
MCC = 0.67
LocError = 6.0 mm
WaveError = 3.7 µV
MCC = 0.79
MIA
LocError = 1.4 mm
WaveError = 2.3 µV
MCC = 1.00
LocError = 2.5 mm
WaveError = 3.0 µV
MCC = 0.91
LocError = 5.7 mm
WaveError = 3.4 µV
MCC = 0.87
Miss True Location
Hit True Location
False Alarm Beamformer Location
Hit Beamformer Location

Examples of localization performances (MCC values) for Search Method by Source number (4, 6, and 8) for Monte-Carlo run #1 with an SNR = 0.2 and intra-pair correlation = 0.6. True source locations are colored as green baskets if they had a corresponding Hit for an observed source within 15 mm. Localization Error (LocError), waveform error (WaveError), and localization performance (Mathews Correlation Coefficient; MCC) for each condition are listed in parentheses above the images. MCC equal to 1 represents perfect localization performance, 0 represents chance performance, and -1 represents complete failure.

Localization Performance

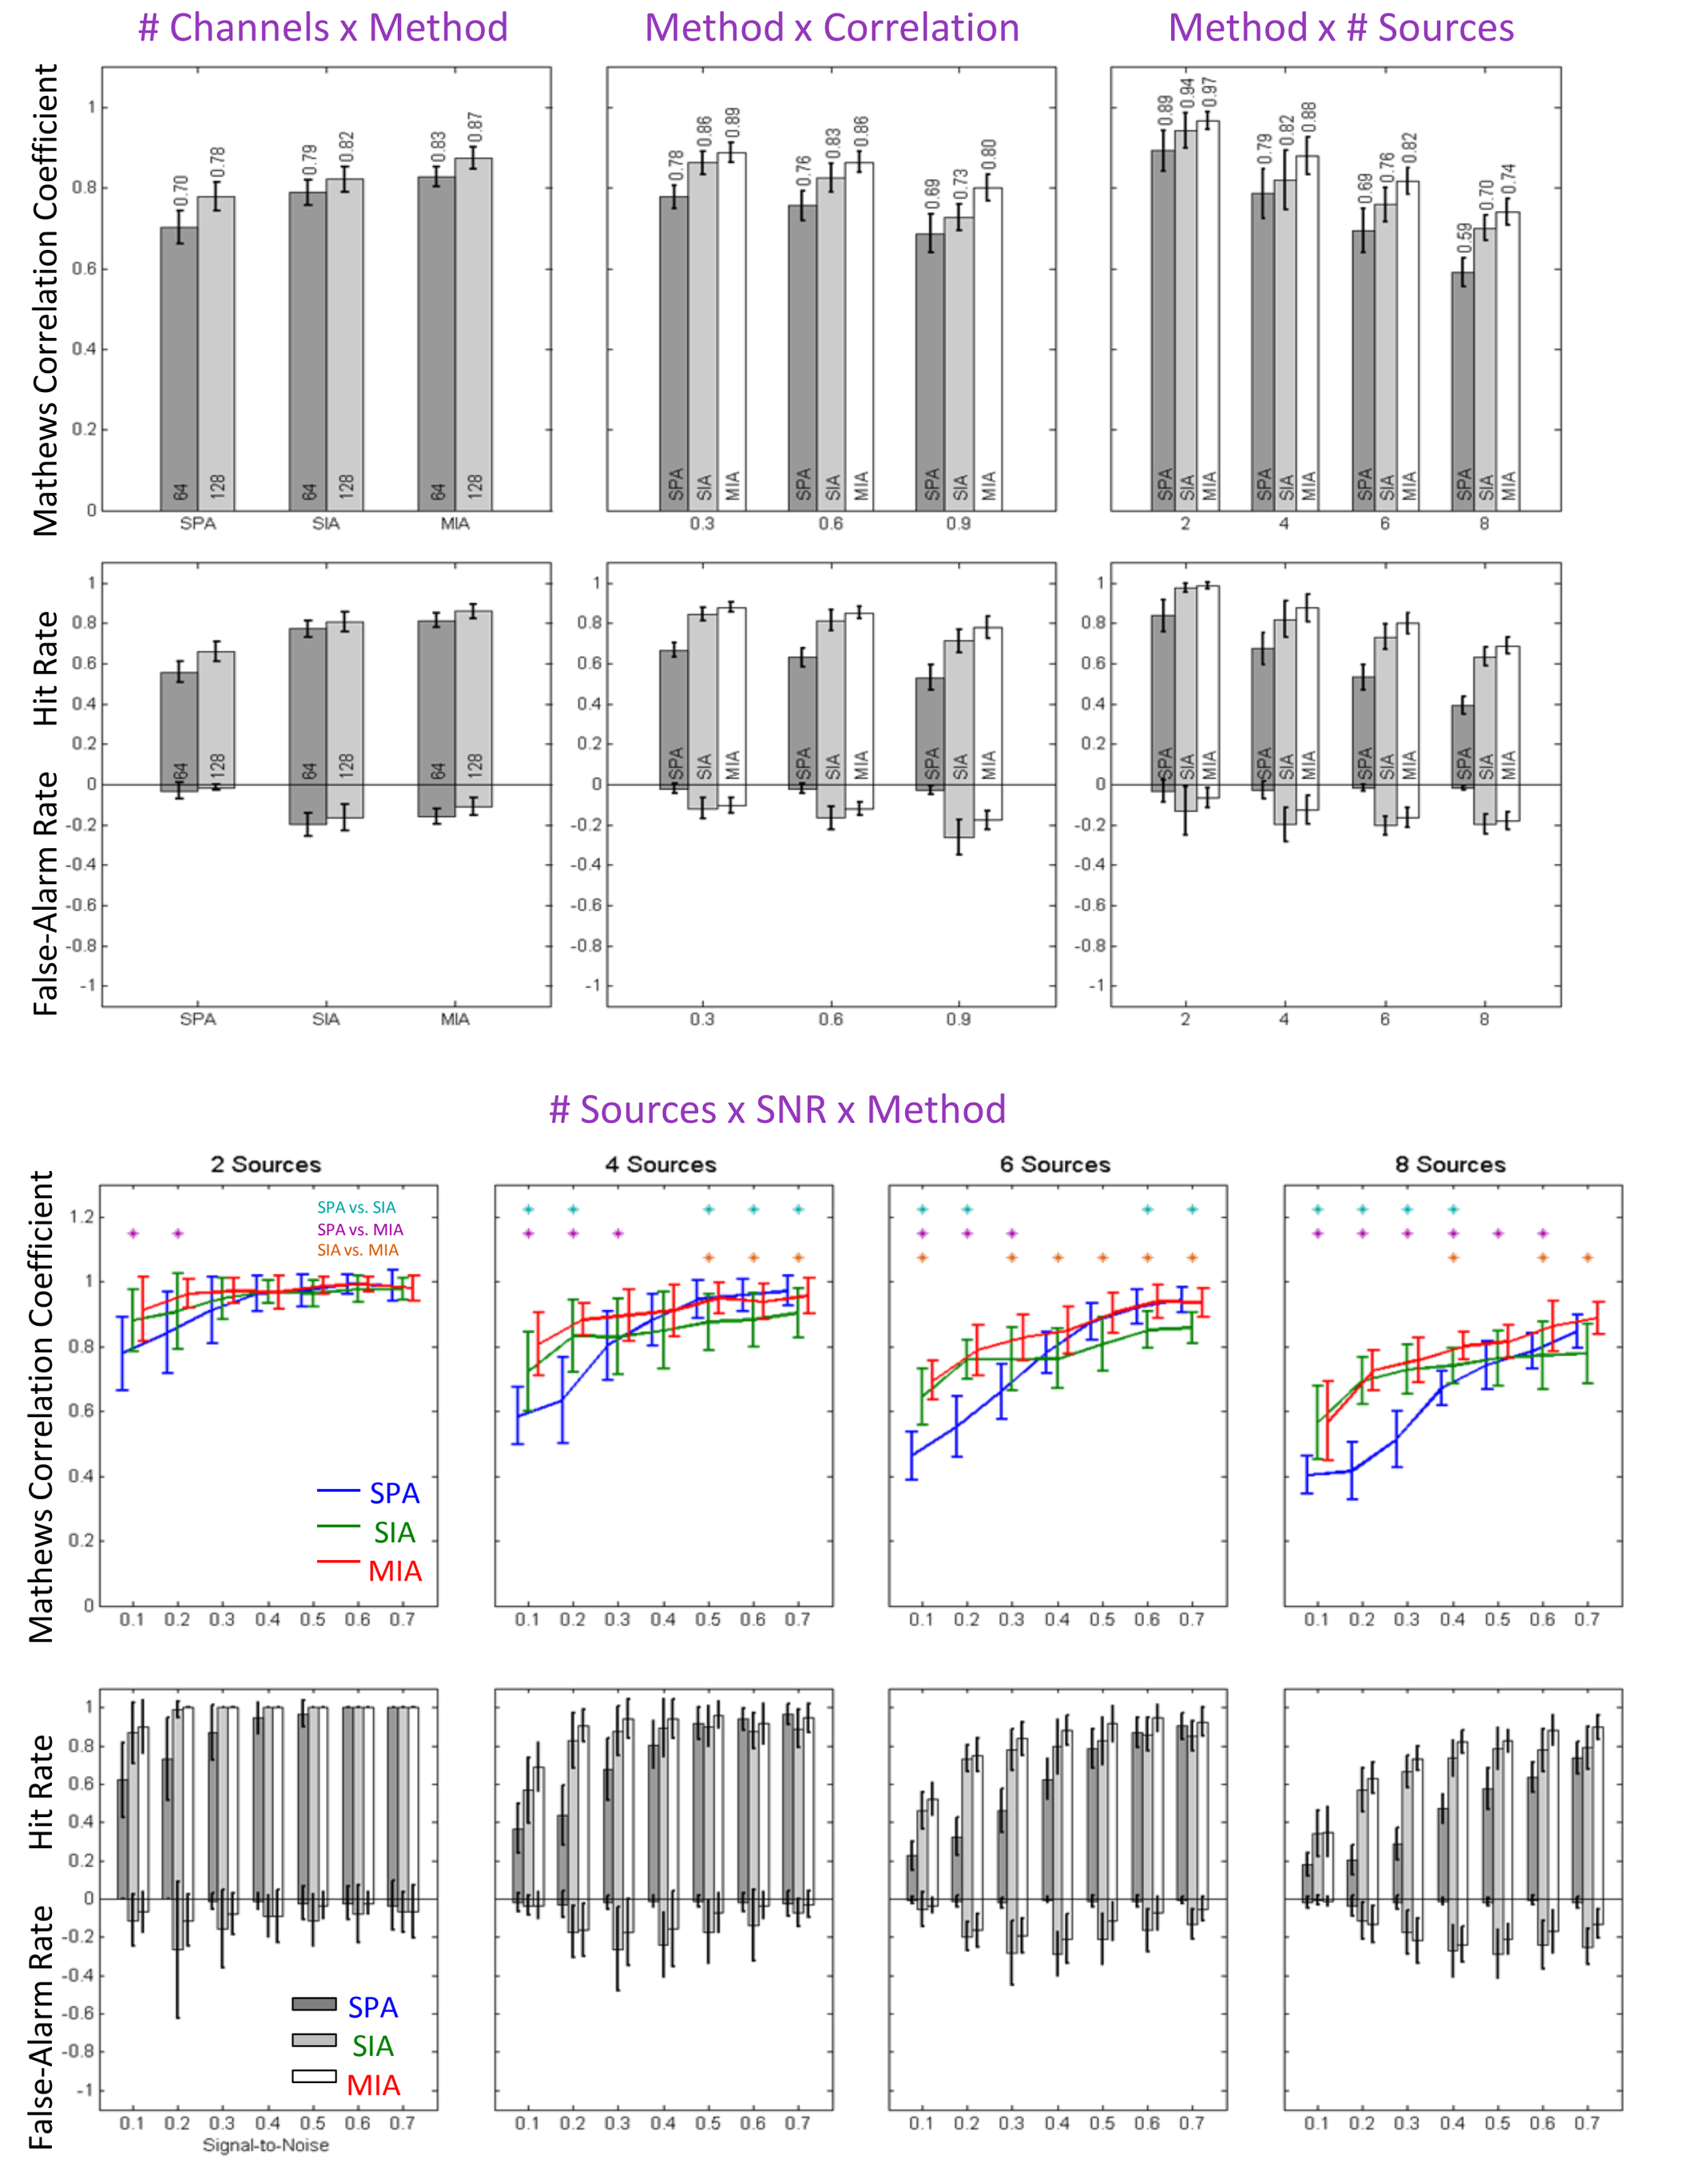
Channels x Method
Method x Correlation
Method x # Sources
Mathews Correlation Coefficient
Hit Rate
False-Alarm Rate
Sources x SNR x Method
2 Sources
4 Sources
6 Sources
8 Sources
SPA vs. SIA
SPA vs. MIA
SIA vs. MIA
SPA
SIA
MIA
Signal-to-Noise

Localization Error

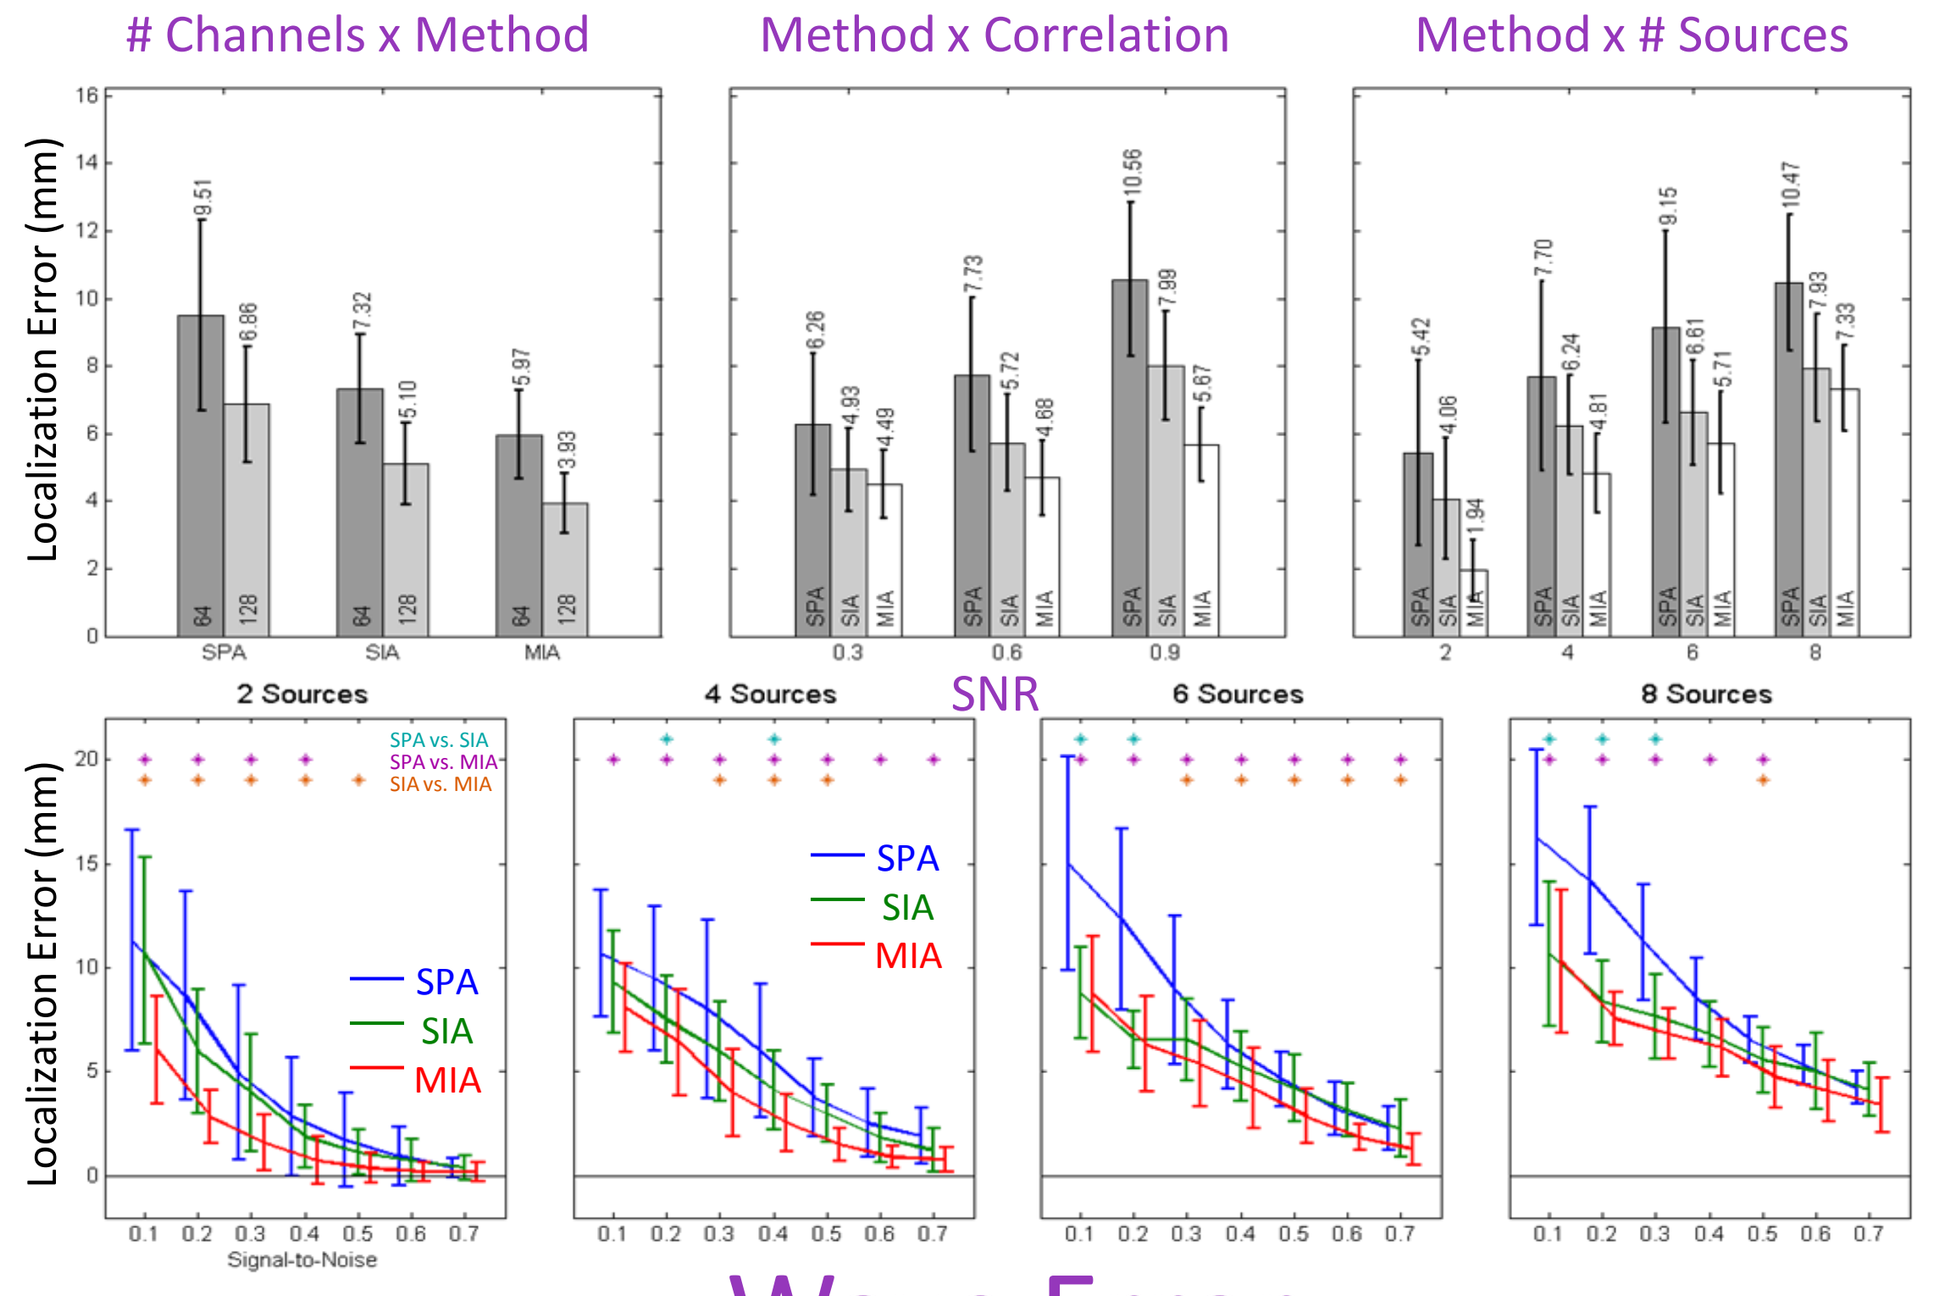
Channels x Method
Method x Correlation
Method x # Sources
Localization Error (mm)
2 Sources
4 Sources
SNR
6 Sources
8 Sources
SPA vs. SIA
SPA vs. MIA
SIA vs. MIA
SPA
SIA
MIA
Signal-to-Noise

Wave Error

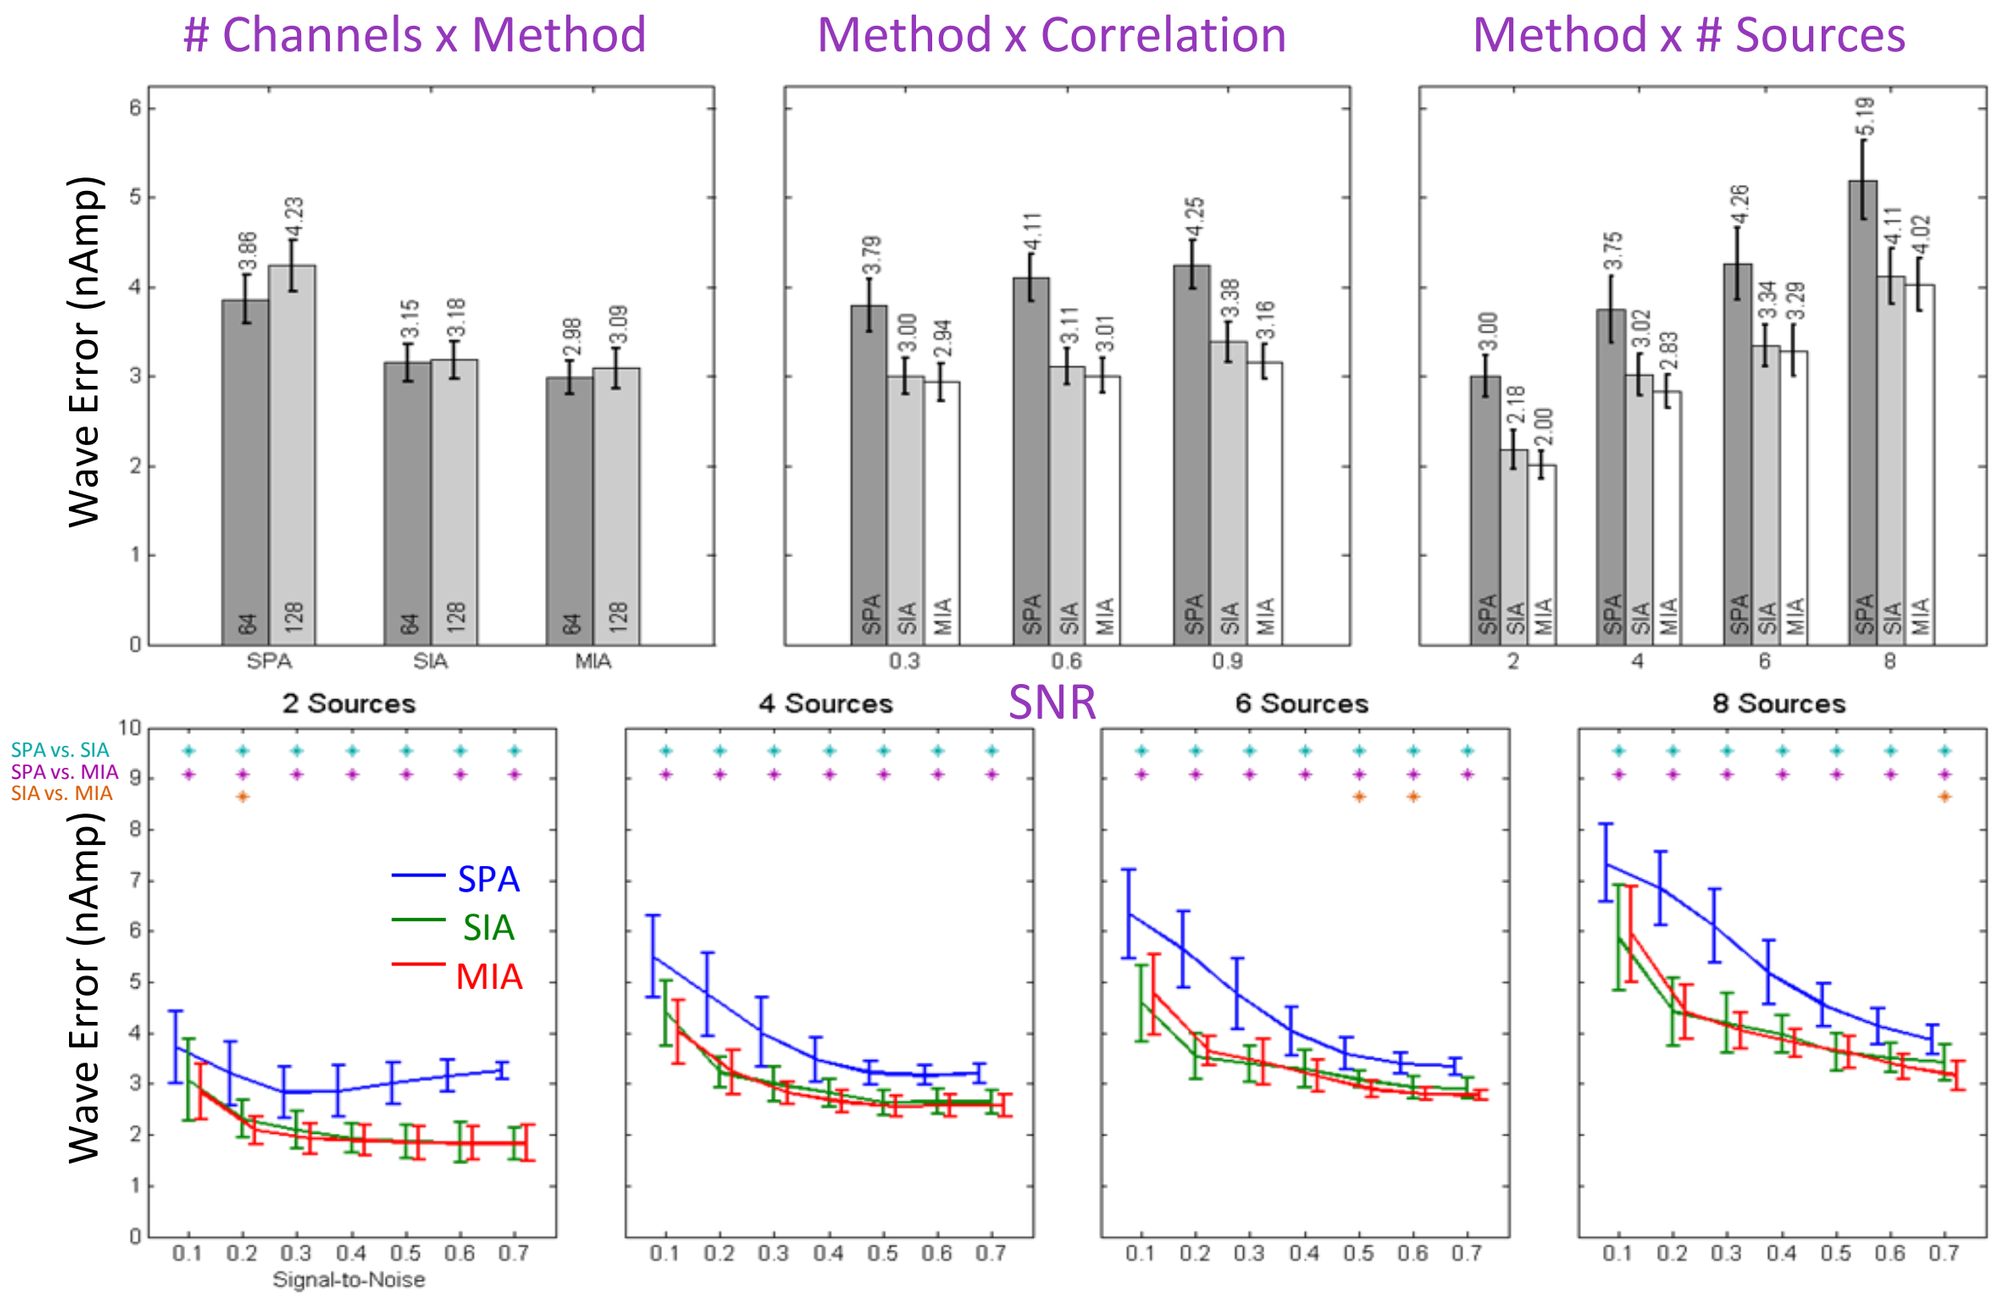
Channels x Method
Method x Correlation
Method x # Sources
Wave Error (nAmp)
2 Sources
4 Sources
SNR
6 Sources
8 Sources
SPA vs. SIA
SPA vs. MIA
SIA vs. MIA
SPA
SIA
MIA
Signal-to-Noise

Functional Connectivity Challenge

WATCH OUT for Leaks!

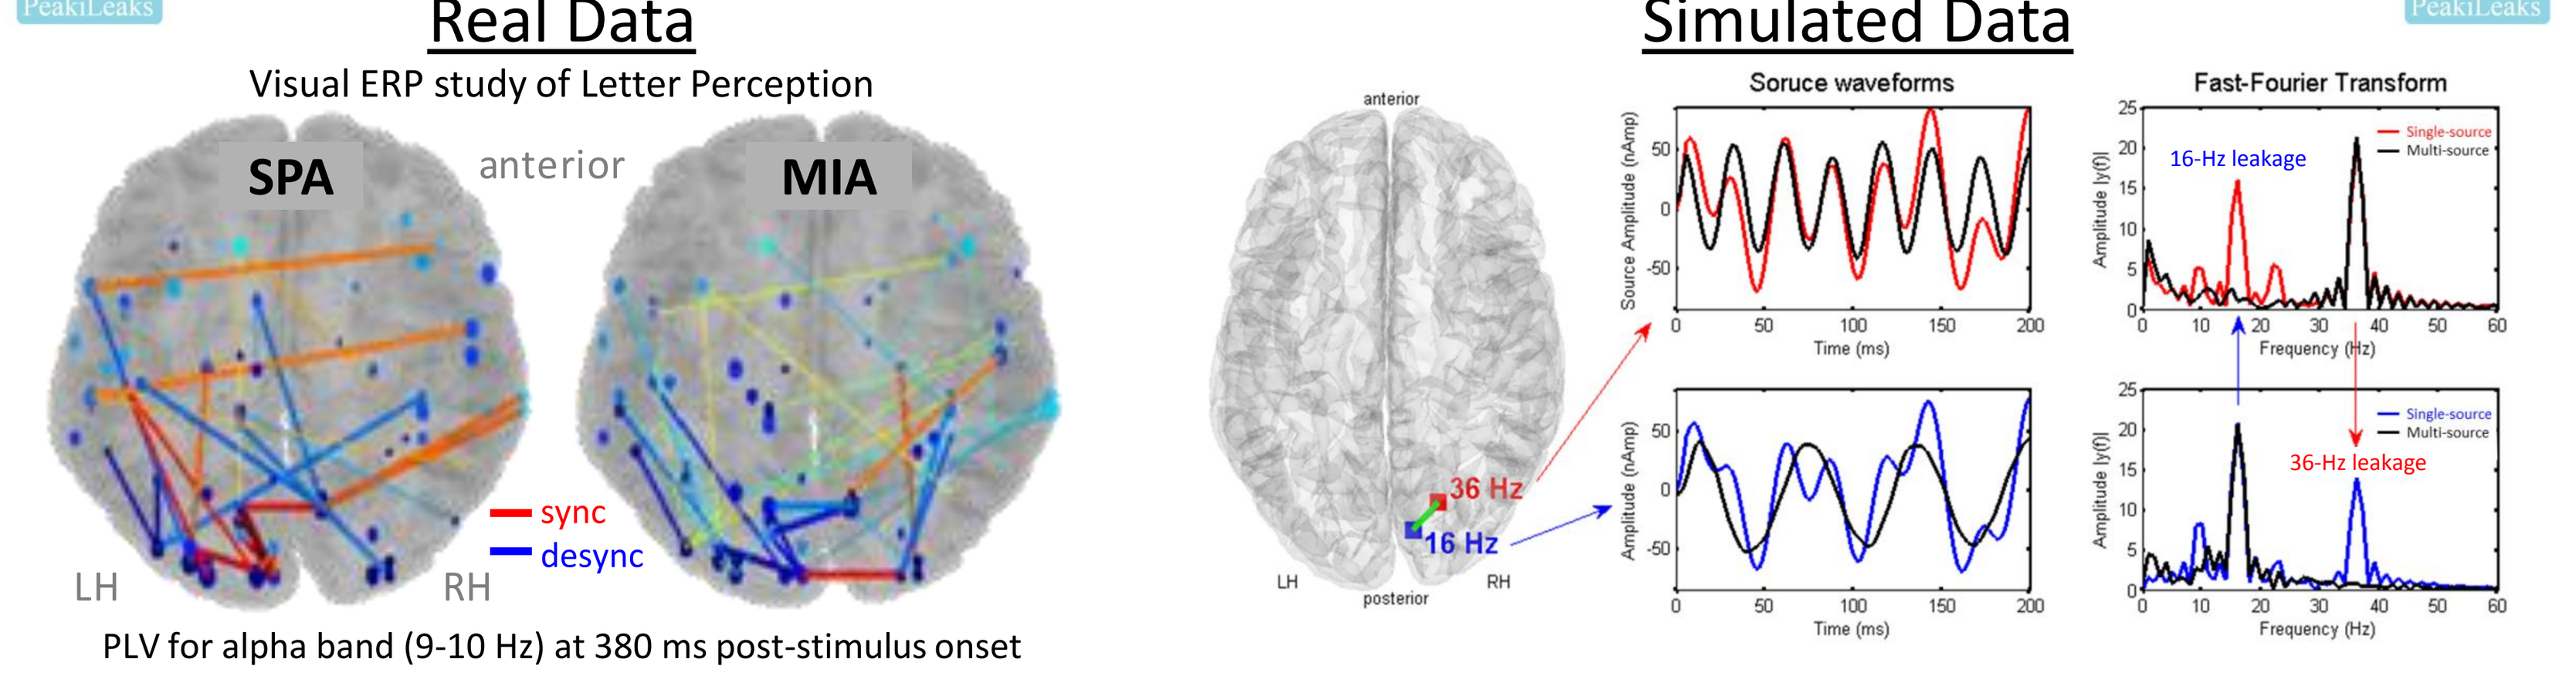
Real Data
Visual ERP study of Letter Perception
SPA
anterior
MIA
sync
desync
LH
RH
PLV for alpha band (9-10 Hz) at 380 ms post-stimulus onset
Simulated Data
anterior
posterior
36 Hz
16 Hz
Soruce waveforms
Fast-Fourier Transform
16-Hz leakage
36-Hz leakage
Single-source
Multi-source
Time (ms)
Frequency (Hz)

Type I Errors: Single-Source Beamformer & Phase-Locking Value (PLV)

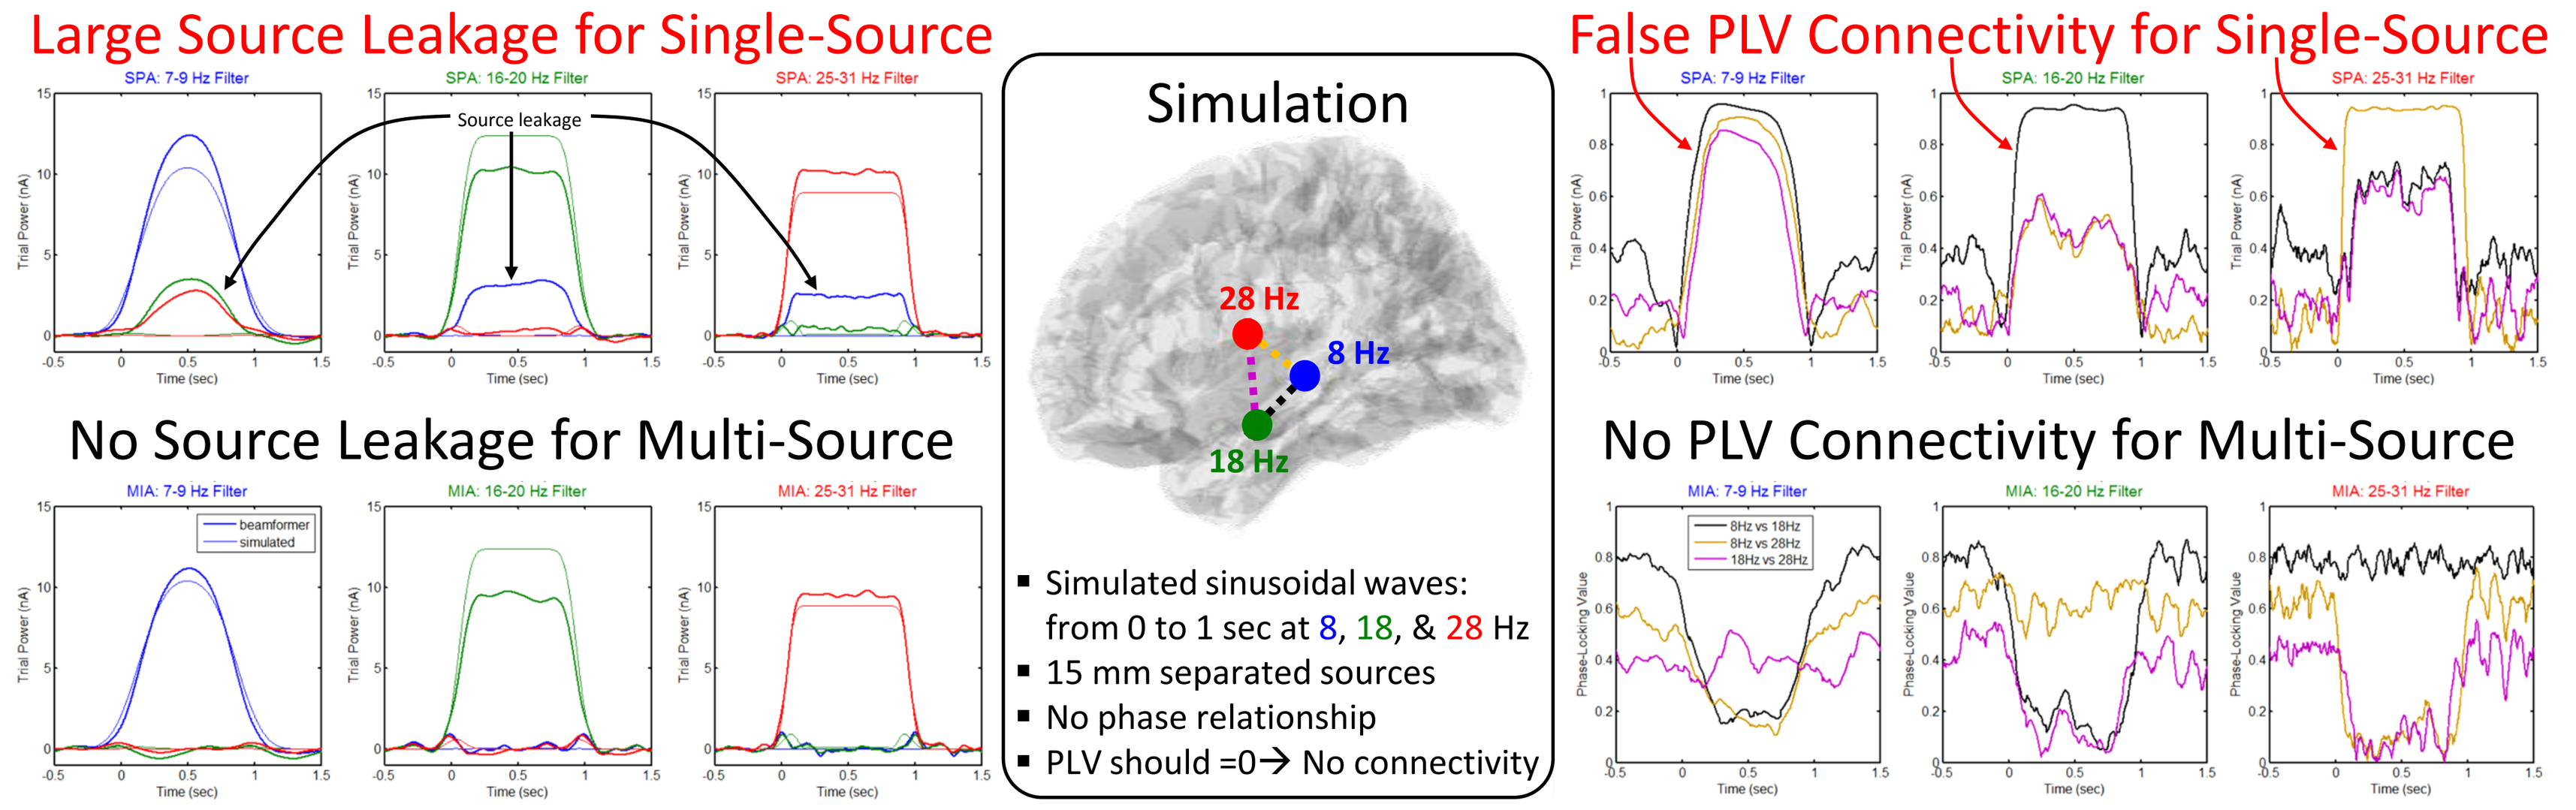
Large Source Leakage for Single-Source
Source leakage
SPA: 7-9 Hz Filter
SPA: 16-20 Hz Filter
SPA: 25-31 Hz Filter
No Source Leakage for Multi-Source
MIA: 7-9 Hz Filter
MIA: 16-20 Hz Filter
MIA: 25-31 Hz Filter
Simulation
28 Hz
8 Hz
18 Hz
False PLV Connectivity for Single-Source
No PLV Connectivity for Multi-Source
8Hz vs 18Hz
8Hz vs 28Hz
18Hz vs 28Hz

- Simulated sinusoidal waves: from 0 to 1 sec at 8, 18, & 28 Hz
- 15 mm separated sources
- No phase relationship
- PLV should =0→ No connectivity

Type II Errors: Single-Source Beamformer & Phase-Lag Index (PLI)

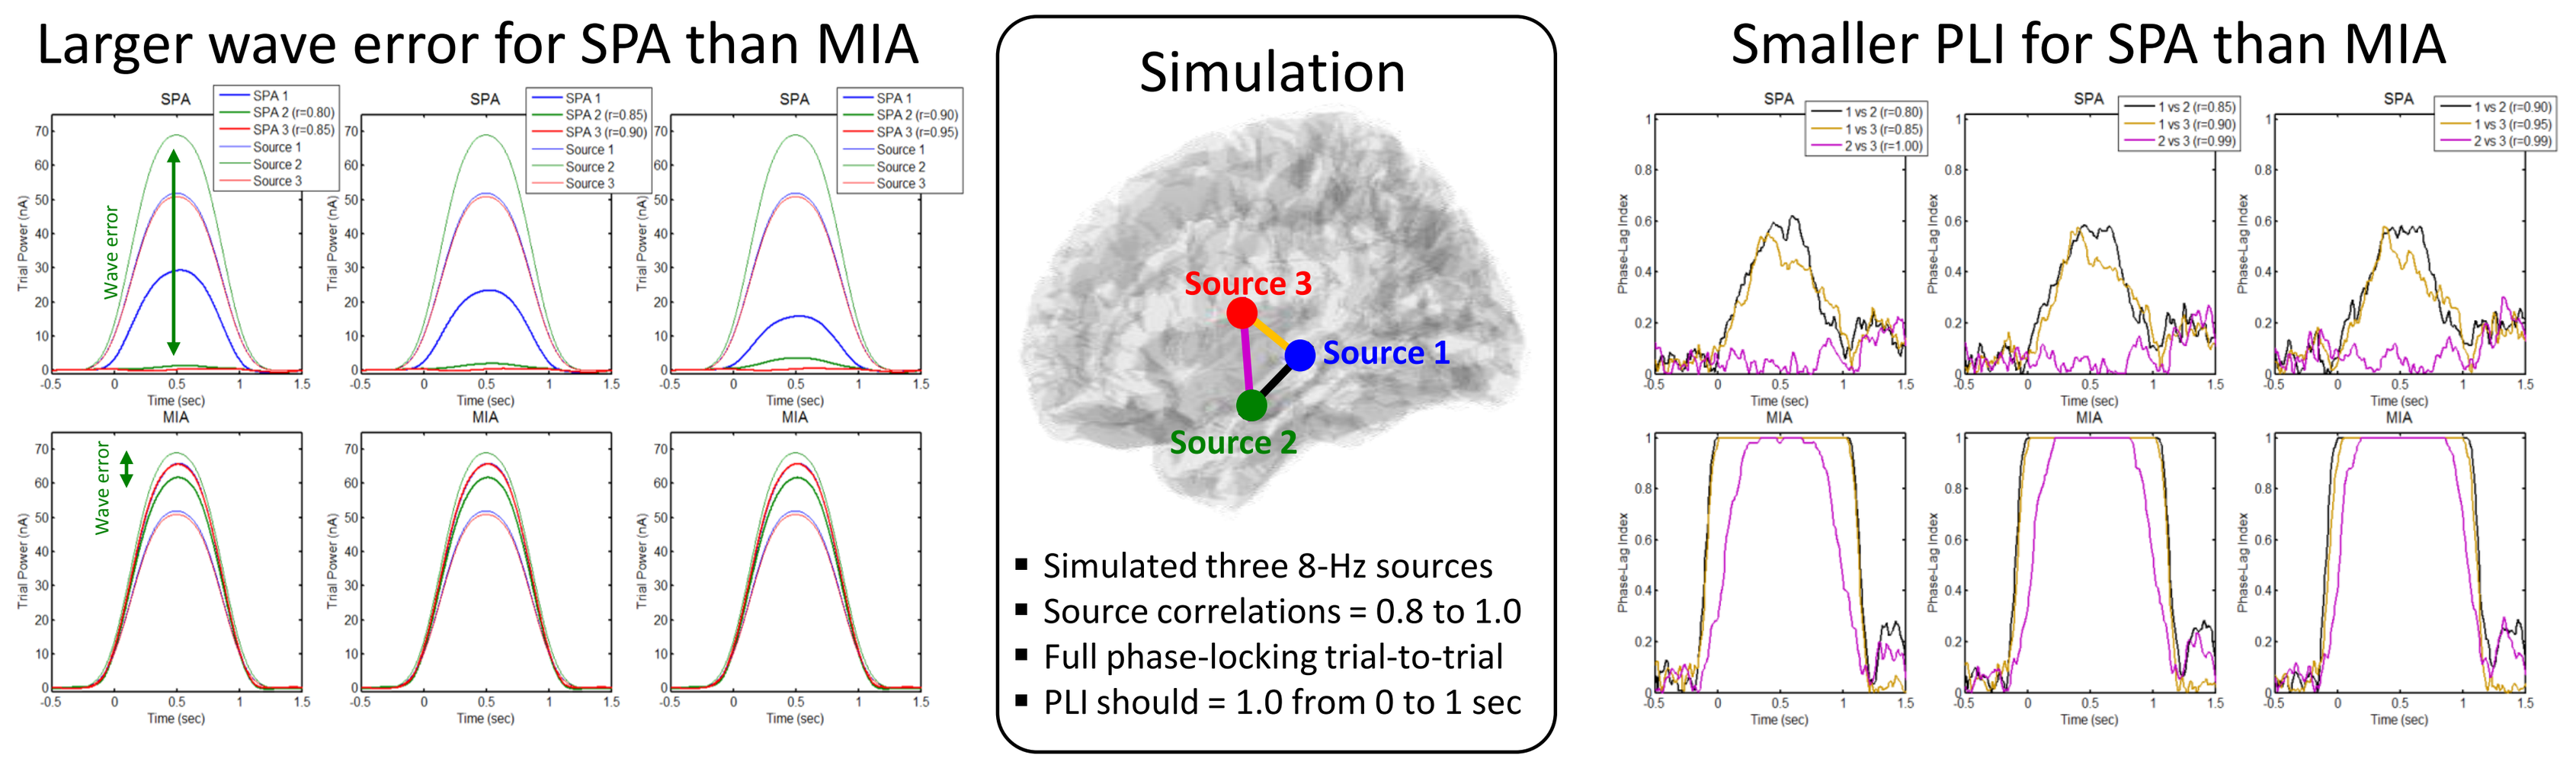
Larger wave error for SPA than MIA
Wave error
SPA
MIA
Simulation
Source 3
Source 1
Source 2
Smaller PLI for SPA than MIA
Time (sec)

- Simulated three 8-Hz sources
- Source correlations = 0.8 to 1.0
- Full phase-locking trial-to-trial
- PLI should = 1.0 from 0 to 1 sec

Conclusion

1) Use Multi-Source Beamformer (MIA) for ERP analyses in order to improve
 - localization performance
 - localization precision
 - source waveform reconstruction
 - finding “true” connectivity among sources
2) Source leakage can lead to finding false connections (Type I error)
3) Compensating for source leakage using PLI can lead to reduction in “true” connectivity (Type II error) when sources are highly correlated (e.g., bilateral auditory activation r>0.9)

Acknowledgements

Support and funding for this project were provided by: NSERC CRSNG, Canada Foundation for Innovation / Fondation canadienne pour l'innovation, UBC

a place of mind — THE UNIVERSITY OF BRITISH COLUMBIA

Available @ www.audiospeech.ubc.ca/research/branelab/